Abstracts / Osteoarthritis and Cartilage 20 (2012) S54–S296 S287

572

APPA PROVIDES SYMPTOM RELIEF IN CLINICAL CANINE OSTEOARTHRITIS

S. Glasson [1], N. Larkins [2]. [1] *ALK Intl., Cook, Australia;* [2] *ALK Intl., Barcelona, Spain*

Purpose: Lameness due to osteoarthritis (OA) is common in aged dogs and altered weight bearing can be objectively quantified. Oral APPA (apocynin and paeonol), and underwater treadmill therapy (UTW) were evaluated in client-owned dogs with OA, for normalization of weight bearing utilizing force-plate measurements, pre and post 4 weeks of treatment.

Methods: OA was diagnosed in client-owned dogs by physical examination and confirmed by radiography. Thirty-two dogs were enrolled with half having elbow and half hip OA. Treatment consisted of either 50 mg/kg BID oral APPA or twice weekly sessions of underwater treadmill (UWT) therapy for 4 weeks with all dogs receiving both treatments, with a wash-out period of 3 weeks in-between. Half the animals received APPA first, and half the animals received UWT first. Force plate measures were obtained before and after each treatment at a consistent treadmill speed for each dog (average of 0.6 $\pm$ 0.09 m/s). No other pain medications, including nutraceuticals, were administered to any of the dogs for the duration of the study. Ground reaction force (GRF) measures from 5 strides were used to calculate peak vertical force (PFz), mean vertical force (MFz), and vertical impulse (IFz). Symmetry indices (SI) for the limb pairs (left and right forelimbs for dogs with elbow OA, left and right hindlimbs for dogs with hip OA) were calculated for each of the three measurements, with a high SI indicating more asymmetry (or lameness), and absolute symmetry giving a SI of zero. Individual dog pre- and post-treatment SI values were compared with Wilcoxon matched pairs non-parametric test.

Results: The dogs enrolled in the study were middle to older-aged and mid-large sized (6 $\pm$ 4 years and 30 $\pm$ 9 kg). Two dogs in the initial UWT group were withdrawn from the study, with one requiring NSAID therapy for pain, and the other had a cardiac tumor. The UWT was well tolerated and the duration of each twice-weekly session steadily increased from 5 $\pm$ 2 min to 21 $\pm$ 7 min over the 4 week treatment period. Twice daily oral APPA was well tolerated for 4 weeks in all but one of the 32 dogs who was removed from the study following mild gastrointestinal signs (vomiting following dosing). A total of 29 dogs completed the study and received the two, 4-week treatments, with a total study length of 11 weeks (including the 3 week wash-out interval). APPA decreased Peak Vertical Force SI when compared to pre-treatment levels ($p < 0.02$, Wilcoxon test), decreased Mean Vertical Force SI ($p = 0.0001$), and decreased Vertical Impulse SI ($p < 0.0001$). Gait symmetry indices following both APPA and UWT approached those obtained historically from healthy dogs.

Conclusions: APPA and UWT therapy were well-tolerated in client-owned dogs and normalized gait asymmetry, as measured with an objective force plate system. APPA normalized gait symmetry indices without the need for UWT equipment or specialized supervision. APPA has also demonstrated decreased disease progression in a rat meniscal tear model of OA, but disease progression was not examined in this dog study. APPA is being further investigated for its ability to decrease pain, clinically in human and veterinary patients, and to decrease cartilage destruction in preclinical models.

573

A RANDOMIZED CONTROLLED STUDY FOR THE COMPARISON OF EFFICACY AND SAFETY ASSESSMENT OF INTRA-ARTICULAR INJECTION OF HIGH MOLECULAR WEIGHT HYALURONIC ACID AND ORAL NON-STEROIDAL ANTI-INFLAMMATORY DRUGS FOR JAPANESE PATIENTS WITH KNEE OSTEOARTHRITIS (UMIN000001026)

M. Ishijima [1], T. Nakamura [2], K. Shimizu [3], K. Hayashi [4], K. Kaneko [1]. For Research Group of Cartilage Metabolism [1] *Juntendo Univ. Graduate Sch. of Med., Tokyo, Japan;* [2] *Univ. of Occupational and Environmental Hlth., Fukuoka, Japan;* [3] *Gifu Univ., Sch. of Med., Gifu, Japan;* [4] *Gunma Univ., Gunma, Japan*

Purpose: Among the pharmacological treatment for knee osteoarthritis (OA), oral non-steroidal anti-inflammatory drugs (NSAIDs) are recommended to be used in OARSI recommendations for the management of OA. Although intra-articular injections of hyaluronic acid (IA-HA) are also recommended, there was very considerable heterogeneity of outcomes between trials. IA-HA is more common in clinical medicine in Japan. However, there is no direct comparison in terms of the efficacy and safety of these two treatments in Japanese patients with knee OA. The aim of this multicenter randomized controlled head-to-head comparison study was to clarify that IA-HA was not inferior to NSAID for the treatment of Japanese patients with knee OA.

Methods: A total of 200 patients with knee OA (K/L grade 1 to 3) were registered from nineteen hospitals and randomized to NSAID (loxoprofen sodium, Loxonin®) or IA-HA (High molecular weight 2,700 kDa HA, Suvenyl®). For patients treated with NSAID, they used NSAID 3 tablets (180 mg)/day for five weeks. For patients treated with IA-HA, intra-articular injection of high molecular weight HA was conducted for 5 times with one week interval. The trial was conducted in accordance with the Declaration of Helsinki. All patients provided written informed consent before enrollment in this trial. The protocol was reviewed by the institutional review board of each institution. The primary endpoint was the percent changes of Japanese Knee Osteoarthritis Measure (JKOM), which is a patient-oriented outcome measure for Japanese patients with knee OA, during 5 weeks observation. The secondary endpoint was the percent differences of pain visual analogue scale (VAS) score. The authors analyzed the full analysis set (FAS) as the primary analysis, based on the concept of intention-to-treat (ITT) and the per protocol set (PPS) as the secondary analysis. As the results obtained by FAS were similar to those by PPS analysis, data obtained from FAS analysis will be presented.

Results: During the 5 weeks of examination, while 20.4% of patients with NSAID were withdrawn, 9.2% of patients with IA-HA were withdrawn. These differences for the frequency of withdrawal between NSAID and IA-HA were statistically significant ($p=0.02$). The frequency for adverse events in patients with NSAID and IA-HA was 10.8% and 1.0%, respectively. These differences for the frequency of adverse events between NSAID and IA-HA were statistically significant ($p=0.003$). No significant changes of JKOM sc ore were observed in patients with either NSAID or IA-HA by the treatment ($p=0.18$ and 0.55, respectively). The difference of percent changes of JKOM score between the two intervention arms (primary outcome measure) was -1.34% (90%CI; -7.68 to 5.01), and IA-HA was non-inferior

to NSAID. Pain VAS scores of the patients with NSAID (56.1) and IA-HA (58.9) at baseline were significantly reduced in comparison to those after the treatment (31.0 and 30.3) ($p<0.001$), respectively. The differences of the secondary outcome measure (% change of pain VAS score) between two intervention arms was -5.74% (90%CI; -23.6 to 12.1), and IA-HA was non-inferior to NSAID for pain. In addition, when the patients were divided into two groups (responder or non-responder) by OMERACT-OARSI response criteria, 51.5% of the patients with IA-HA were classified into “responder”, while 49.5% of those with NSAID were “responder”. Again, there were no significant differences of the frequency of “responder” between these two groups. Logistic regression analysis for odds of responders also revealed that the patients with IA-HA were not inferior to those with NSAID (OR 1.00 (95%CI; 0.56-1.78, $p=0.99$)).

Conclusions: This study showed that the efficacy of IA-HA was not inferior in comparison to that of NSAID, and the safety of IA-HA was superior in comparison to that of NSAID for Japanese patients with knee OA.

574

EXPLORATORY ANALYSIS OF OSTEOARTHRITIS PROGRESSION AMONG MEDICATION USERS: DATA FROM THE OSTEOARTHRITIS INITIATIVE

J.B. Driban [1], G.H. Lo [2], C.B. Eaton [3], K.L. Lapane [4], M. Nevitt [5], W.F. Harvey [1], T.E. McAlindon [1]. [1] *Tufts Med. Ctr., Boston, MA, USA;* [2] *Michael E. DeBakey VA Med. Ctr., Baylor Coll. of Med., Houston, TX, USA;* [3] *Ctr. for Primary Care and Prevention, Alpert Med. Sch. of Brown Univ., Pawtucket, RI, USA;* [4] *Dept. of Epidemiology and Community Hlth., Virginia Commonwealth Univ., Richmond, VA, USA;* [5] *Dept. of Epidemiology and Biostatistics at the Univ. of California, San Francisco, San Francisco, CA, USA*

Purpose: There has been limited success exploring disease modifying interventions for knee osteoarthritis (OA) and it would be cost prohibitive to explore a large number of pharmacological interventions in randomized clinical trials. Therefore, we conducted an exploratory analysis of OA

APPA PROVIDES SYMPTOM RELIEF IN CLINICAL CANINE OSTEOARTHRITIS

Sonya Glasson*; Nick Larkins*
*AKL Technologies Ltd

Background

- The incidence of OA in both humans and dogs is increased by trauma as well as obesity, aging and genetic abnormalities[1].
- Dogs with primary OA represent a relevant translational model to evaluate the effects of new OA analgesics.

Aim

- To test the ability of a investigational oral therapy (APPA) to normalize lameness in dogs with primary OA via an objective force plate method[2].

Methods

- OA was diagnosed by physical examination and confirmed by radiography.
- A total of thirty-two dogs were enrolled in an ethics-approved protocol at School of Veterinary Medicine, University of Vienna, Austria, with half having elbow and half hip OA. Dogs were middle-aged, older (6 ± 4 years) and mid-large sized (30 ± 9 kg).
- Dogs were randomized to 45 mg/kg BID Oral APPA (apocynin and paeonol), or twice weekly sessions of light massage followed by underwater treadmill therapy (UTW).
- Patient enrolment, study retention and ethical concerns precluded a placebo-control design.
- Wash-out occurred before treatment and for a period of 3 weeks in-between treatment cross-over.

Force Plate Measures

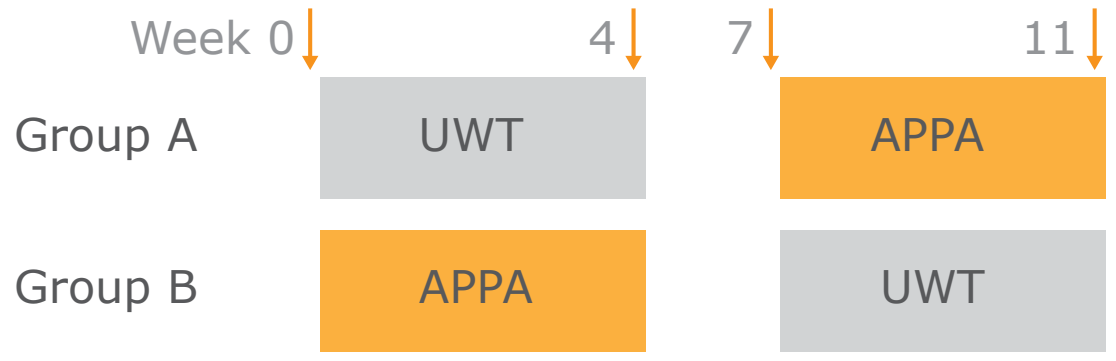
Week 0
4
7
11
Group A
UWT
APPA
Group B
APPA
UWT

- Force plate measures for all 4 limbs were obtained before and after each treatment at a consistent treadmill speed for each dog (average of 0.6 ± 0.09 m/s).
- No other pain medications were administered to any of the dogs for the duration of the study.
- Ground reaction force (GRF) measures from 5 strides were obtained for each leg and normalized to the dog’s body mass and expressed as Newtons per kilogram body mass (N/kg) for:
 - Maximal vertical force (PFz)
 - Mean vertical force (MFz)
 - Vertical Impulse (IFz) = area under the force-timecurve
- Overall symmetry indices (SI) for the limb pairs (left and right fore or hind limbs) were calculated for each of the three measurements.
- $$SI\ \% = 100 \times \left(1 - \frac{\text{Lame P/M/I Force}}{\text{Contralateral P/M/I Force}}\right)$$
- An SI of 0% = perfect symmetry between legs.
- A high SI e.g. > 5% indicates gait asymmetry (lameness).
- Individual dog pre- and post-treatment SI values were compared with Wilcoxon matched pairs non-parametric test.
- Responders were classified as dogs who started with a SI of ≥6%, and had a reduction in ≥6 following treatment.

Results

- Two dogs in the initial UWT group were withdrawn from the study, with one requiring NSAID therapy for pain, and the other had a cardiac tumor.
- The UWT was well tolerated and the duration of each twice-weekly session steadily increased from 5 ± 2 min to 21 ± 7 min over the 4 week treatment period.
- Twice daily oral APPA was well tolerated for 4 weeks in all but one of the 32 dogs who was removed from the study following mild gastrointestinal signs (vomiting).

Dr. Sonya Glasson
sg@aklinternational.com
www.akltechnologies.com

APPA PROVIDES SYMPTOM RELIEF IN CLINICAL CANINE OSTEOARTHRITIS

Sonya Glasson*; Nick Larkins*
*AKL Technologies Ltd

Gait Analysis

- APPA decreased Mean Vertical Force SI ($p < 0.001$) when compared to pre-treatment levels. Gait symmetry indices following treatment approached levels published for normal dogs[3].

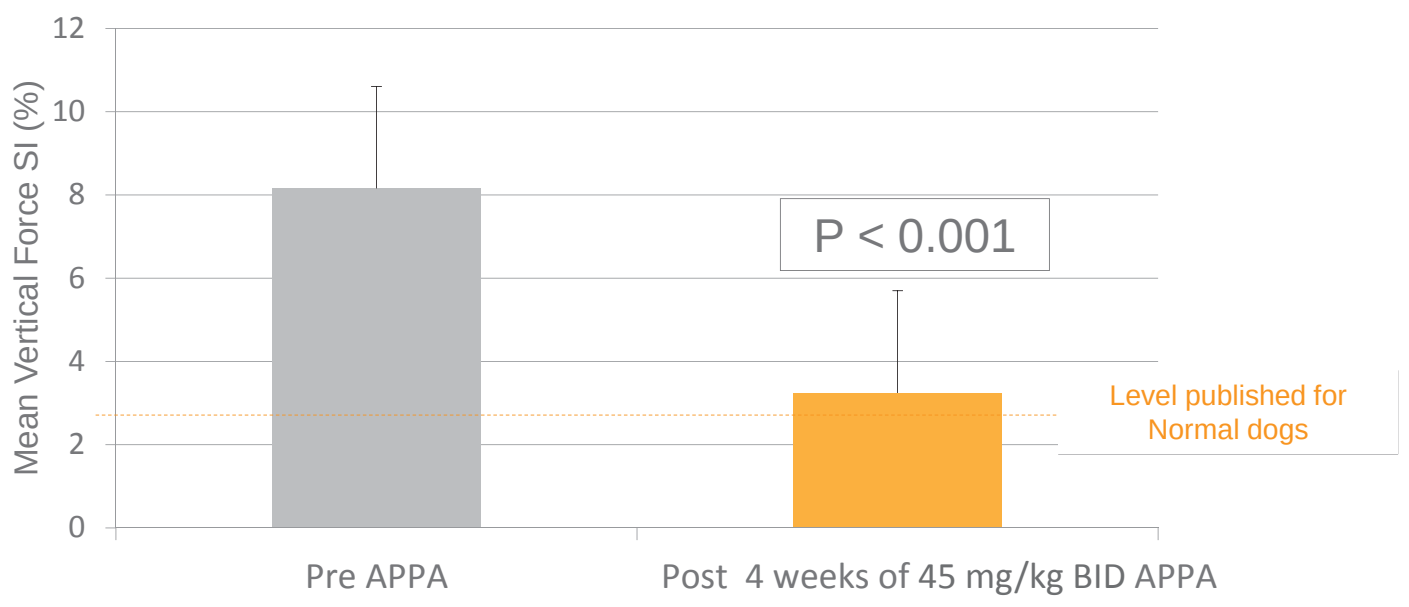
12
10
8
6
4
2
0
Mean Vertical Force SI (%)
P < 0.001
Level published for Normal dogs
Pre APPA
Post 4 weeks of 45 mg/kg BID APPA

- APPA decreased Vertical Impulse SI when compared to pre-treatment levels ($p < 0.0001$).

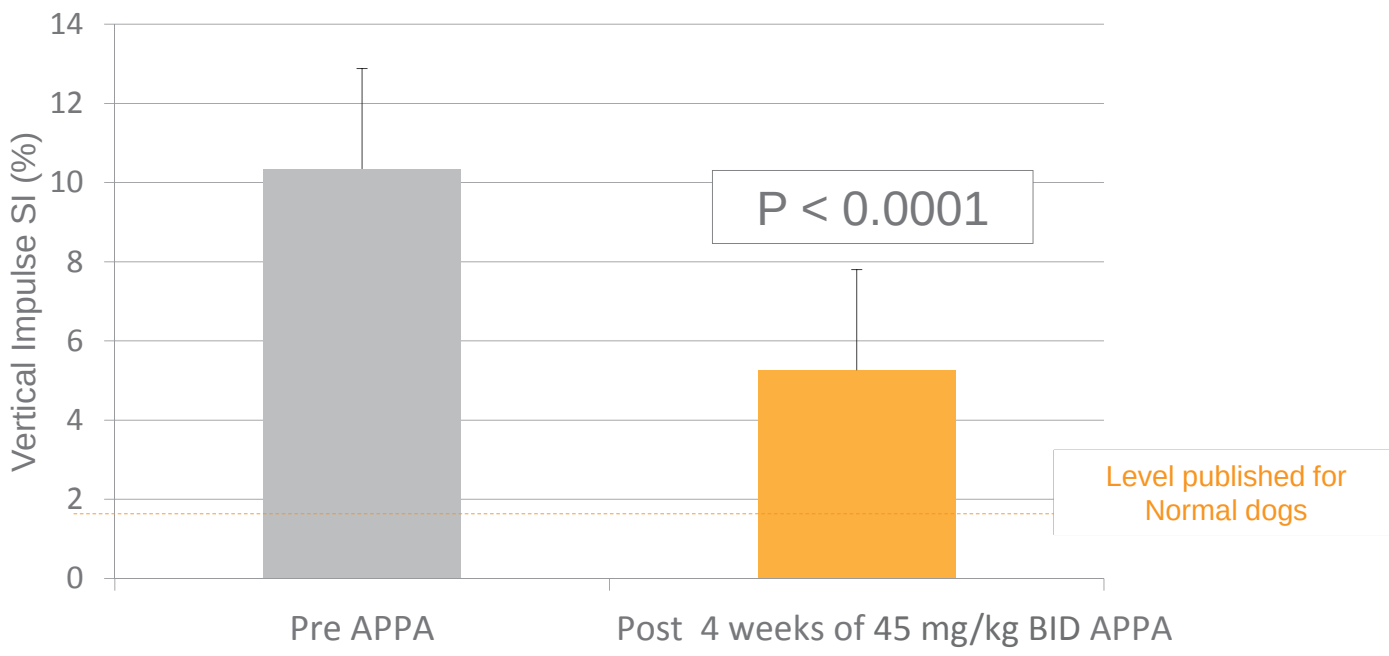
14
12
10
8
6
4
2
0
Vertical Impulse SI (%)
P < 0.0001
Level published for Normal dogs
Pre APPA
Post 4 weeks of 45 mg/kg BID APPA

- APPA decreased Peak Vertical Force SI when compared to pre-treatment levels ($p < 0.02$).

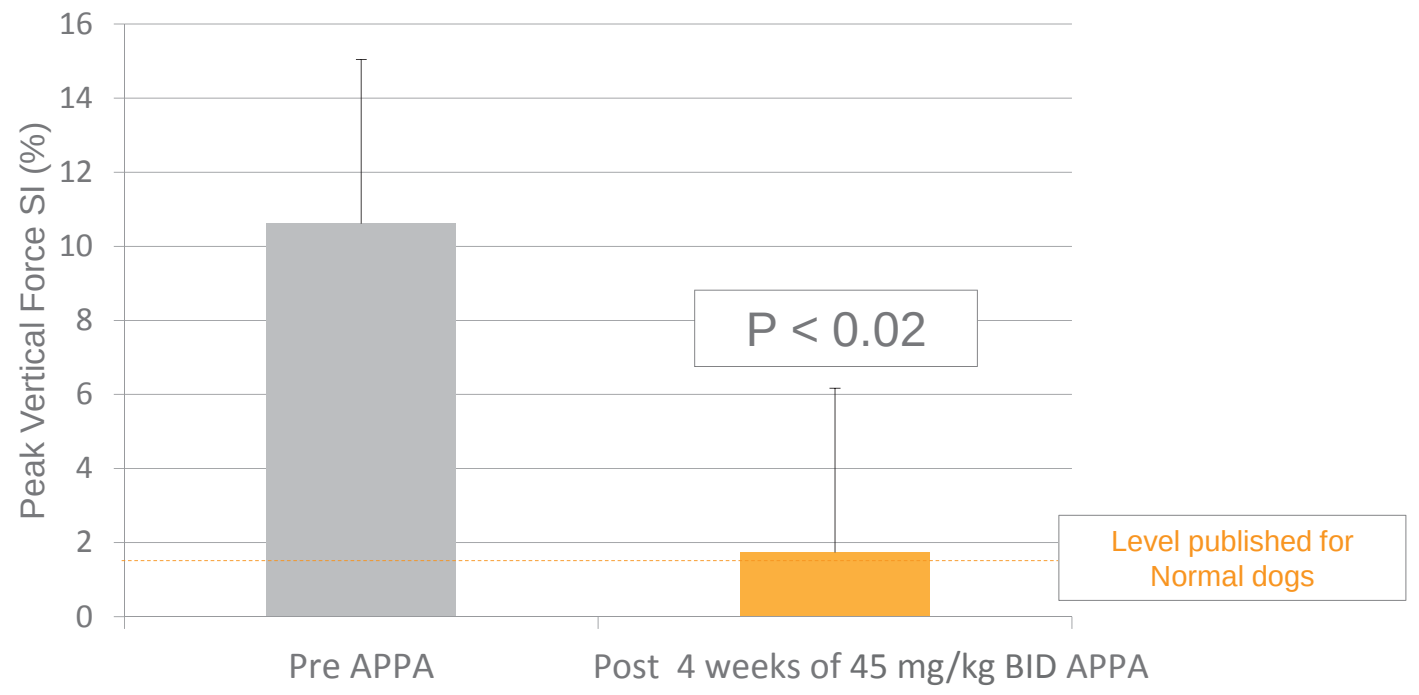
16
14
12
10
8
6
4
2
0
Peak Vertical Force SI (%)
P < 0.02
Level published for Normal dogs
Pre APPA
Post 4 weeks of 45 mg/kg BID APPA

- A high responder rate between 50-80% was observed when comparing individual pre and post-treatment Force Plate measures

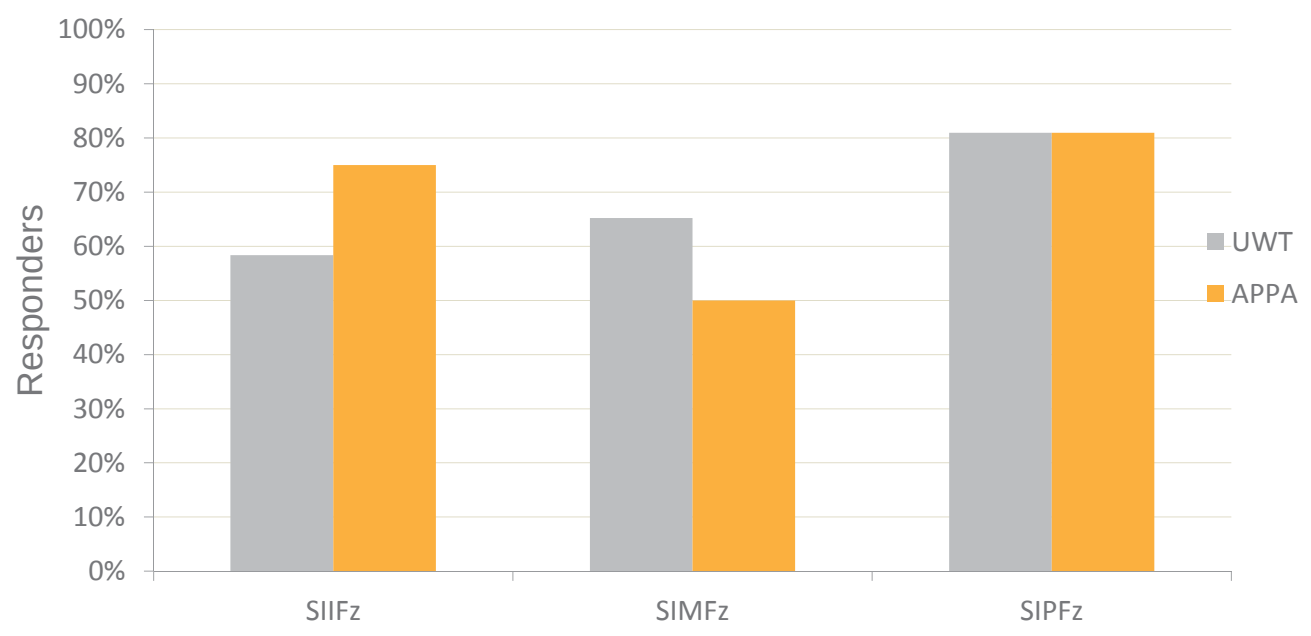
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Responders
UWT
APPA
SIIFz
SIMFz
SIPFz

Conclusions

- APPA and UWT therapy were well-tolerated in client-owned dogs and normalized gait asymmetry, as measured with an objective force plate system.
- APPA normalized gait symmetry indices without the need for UWT equipment or specialized supervision.
- APPA has also demonstrated decreased disease progression in a rat meniscal tear model of OA (see poster # 132).
- APPA is being further investigated for its ability to decrease pain, clinically in human and veterinary patients, and to decrease cartilage destruction in preclinical models.

References

1. Patronek G, Waters D, Glickman L. Comparative longevity of pet dogs and humans, Implications for gerontology research. J Gerontal A Biol Sci Med Sci 1997; 52 B: 171-178.
2. Bochstahler BA et al. Reliability of ground reaction forces measured on a treadmill system in dogs. Veterinary J 2007;173:373-378.
3. Bochstahler BA et al. Compensatory load redistribution in naturally occurring osteoarthritis of the elbow joint and induced weight-bearing lameness of the forelimbs compared with clinically sound dogs. Vet J 2009;180:202-12

Acknowledgements: The Investigators at Vienna University and the Clients who participated in the study.

AKL TECHNOLOGIES LTD
An AKL International Ltd. Company
PO Box 568, Granite House, La Grande Rue, St Martin, Guernsey, GY1 6JS
United Kingdom +44 7929 223575

Dr. Sonya Glasson
sg@aklinternational.com
www.akltechnologies.com