Poster number: 447

EFFECT OF APPA (combination of Apocynin and Paeonol) COMPOUND ON CELLULAR SENESCENCE USING HUMAN ARTICULAR CHONDROCYTES

Mercedes Fernández-Moreno[1], Tamara Hermida-Gómez[1,2], Nicholas Larkins[3], Alan Reynolds[3] and Francisco J. Blanco[1]

1) Servicio de Reumatología. Instituto de Investigación Biomédica de A Coruña (INIBIC). Complexo Hospitalario Universitario de A Coruña (CHUAC), Sergas. Universidade da Coruña (UDC), Spain. 2) Centro de investigación biomédica en Red, Bioingenieria, Biomatereial y Nanomedicina (CIBER-BBN), Spain. 3) AKL Therapeutics, Stevenage Bioscience, United Kingdom.

Purpose Osteoarthritis (OA) is the most common rheumatological illness and a major cause of pain and disability in older adults, but without effective treatment. Senotherapeutics are a class of drugs or natural compounds comprising two members; senomorphics and senolytics. Their main target is to eliminate or delay the adverse effects of cellular senescence and consequently, the process of aging and age-related pathologies such as OA. APPA, a combination of Apocynin (AP) and Paeonol (PA), has shown efficacy in rat and canine models of OA and our previous data showed that APPA has the capacity to modulate several parameters related with OA process in human chondrocytes. ***Aim*** To study the effect of APPA on cellular senescence of human articular chondrocytes.

Methods: Senescence was induced with Etoposide at 2 µM or 20 µM+OSM 10 ng/ml in the chondrocyte cell line TC/28a2 and human chondrocytes from OA hip cartilage respectively. The number of viable cells were evaluated using Hoechst stain and the number of nuclei counted. Senescent cells were evaluated by flow cytometry (FDG) and CDKN1A gene expression by RT-PCR. The susceptibility of cells to apoptosis was analyzed by flow cytometer using Annexin-V and Propidium Iodide (PI). Unpaired Mann Whitney test was used to evaluate differences between groups. Data are presented as the mean ± standard error mean (SEM). Statistical analyses were performed with GraphPad Prism version 8.

Results

In TC/28a2 cells, Etoposide 2 µM increased the number of senescent cells in comparison with basal condition (3.42±0.91 vs 8.56±1.62; p=0.03) (**Figure 1-A**). **APPA 10 µg/ml significantly reduced the number of senescent cells induced by Etoposide** 2 µM (0.99±4.7e^{-5} vs 0.66±0.17; p=0.02) (**Figure 1-B**).

A

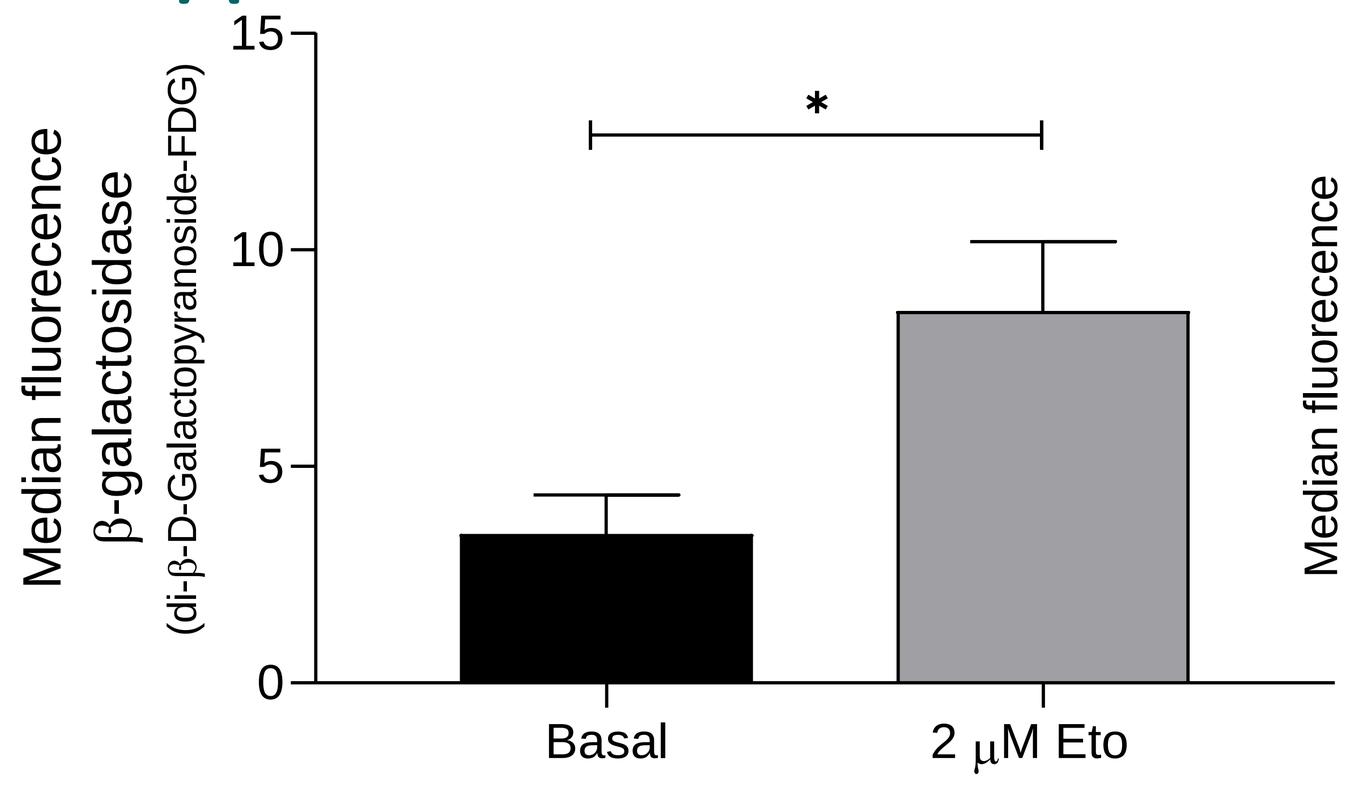
Median fluorecence
β-galactosidase
(di-β-D-Galactopyranoside-FDG)
15
10
5
0
*
Basal
2 µM Eto

B

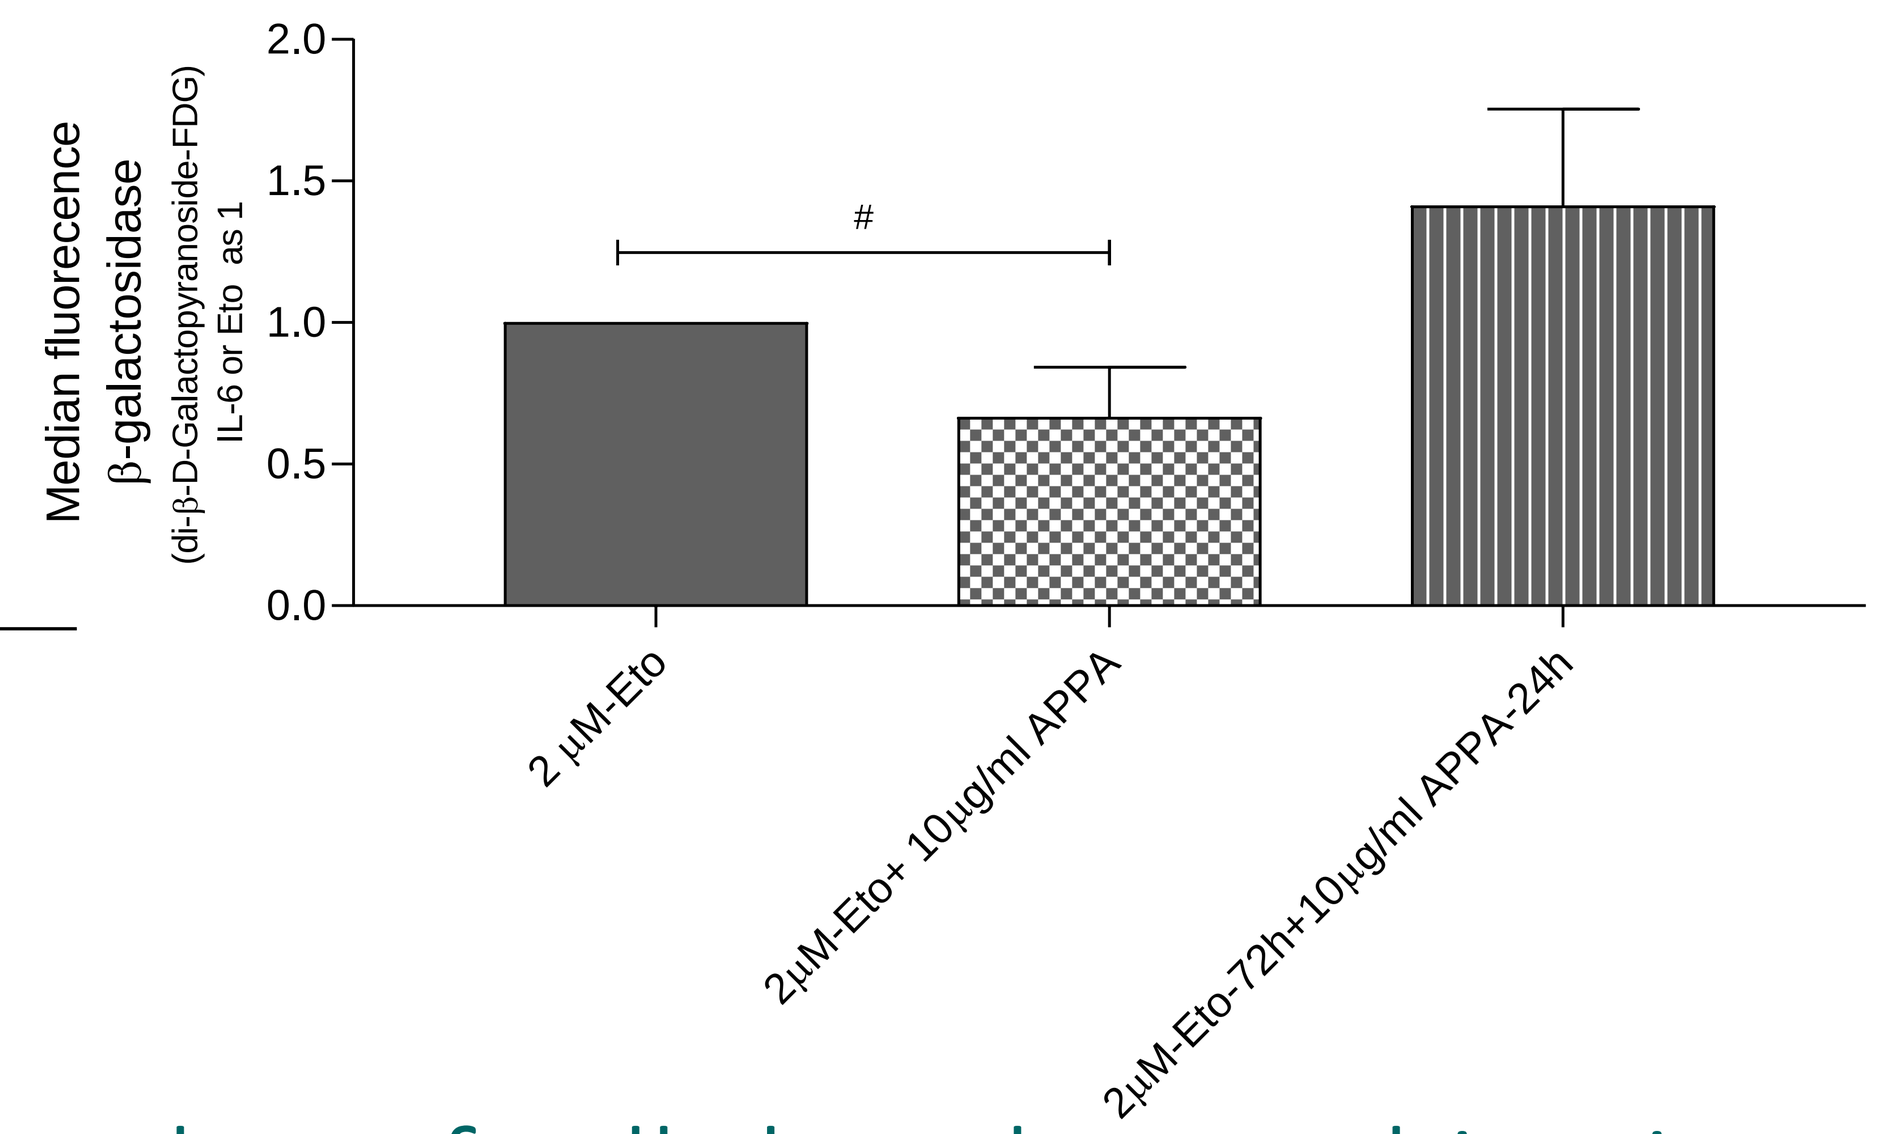
Median fluorecence
β-galactosidase
(di-β-D-Galactopyranoside-FDG)
IL-6 or Eto as 1
2.0
1.5
1.0
0.5
0.0
#
2 µM-Eto
2µM-Eto+ 10µg/ml APPA
2µM-Eto-72h+10µg/ml APPA-24h

Figure 1. Median of fluorescence ß-galactosidase. A) expressed relative to basal condition. **B)** expressed relative to Etoposide condition. *p<0.05; #p<0.05.

Etoposide 2 µM reduced the total number of cells but the combination of **Etoposide with APPA 10 µg/ml increased the number of positive nuclei** (0.99±3.17e^{-5} vs 1.54±0.23, p=0.0062 (**Figure 2**).

A

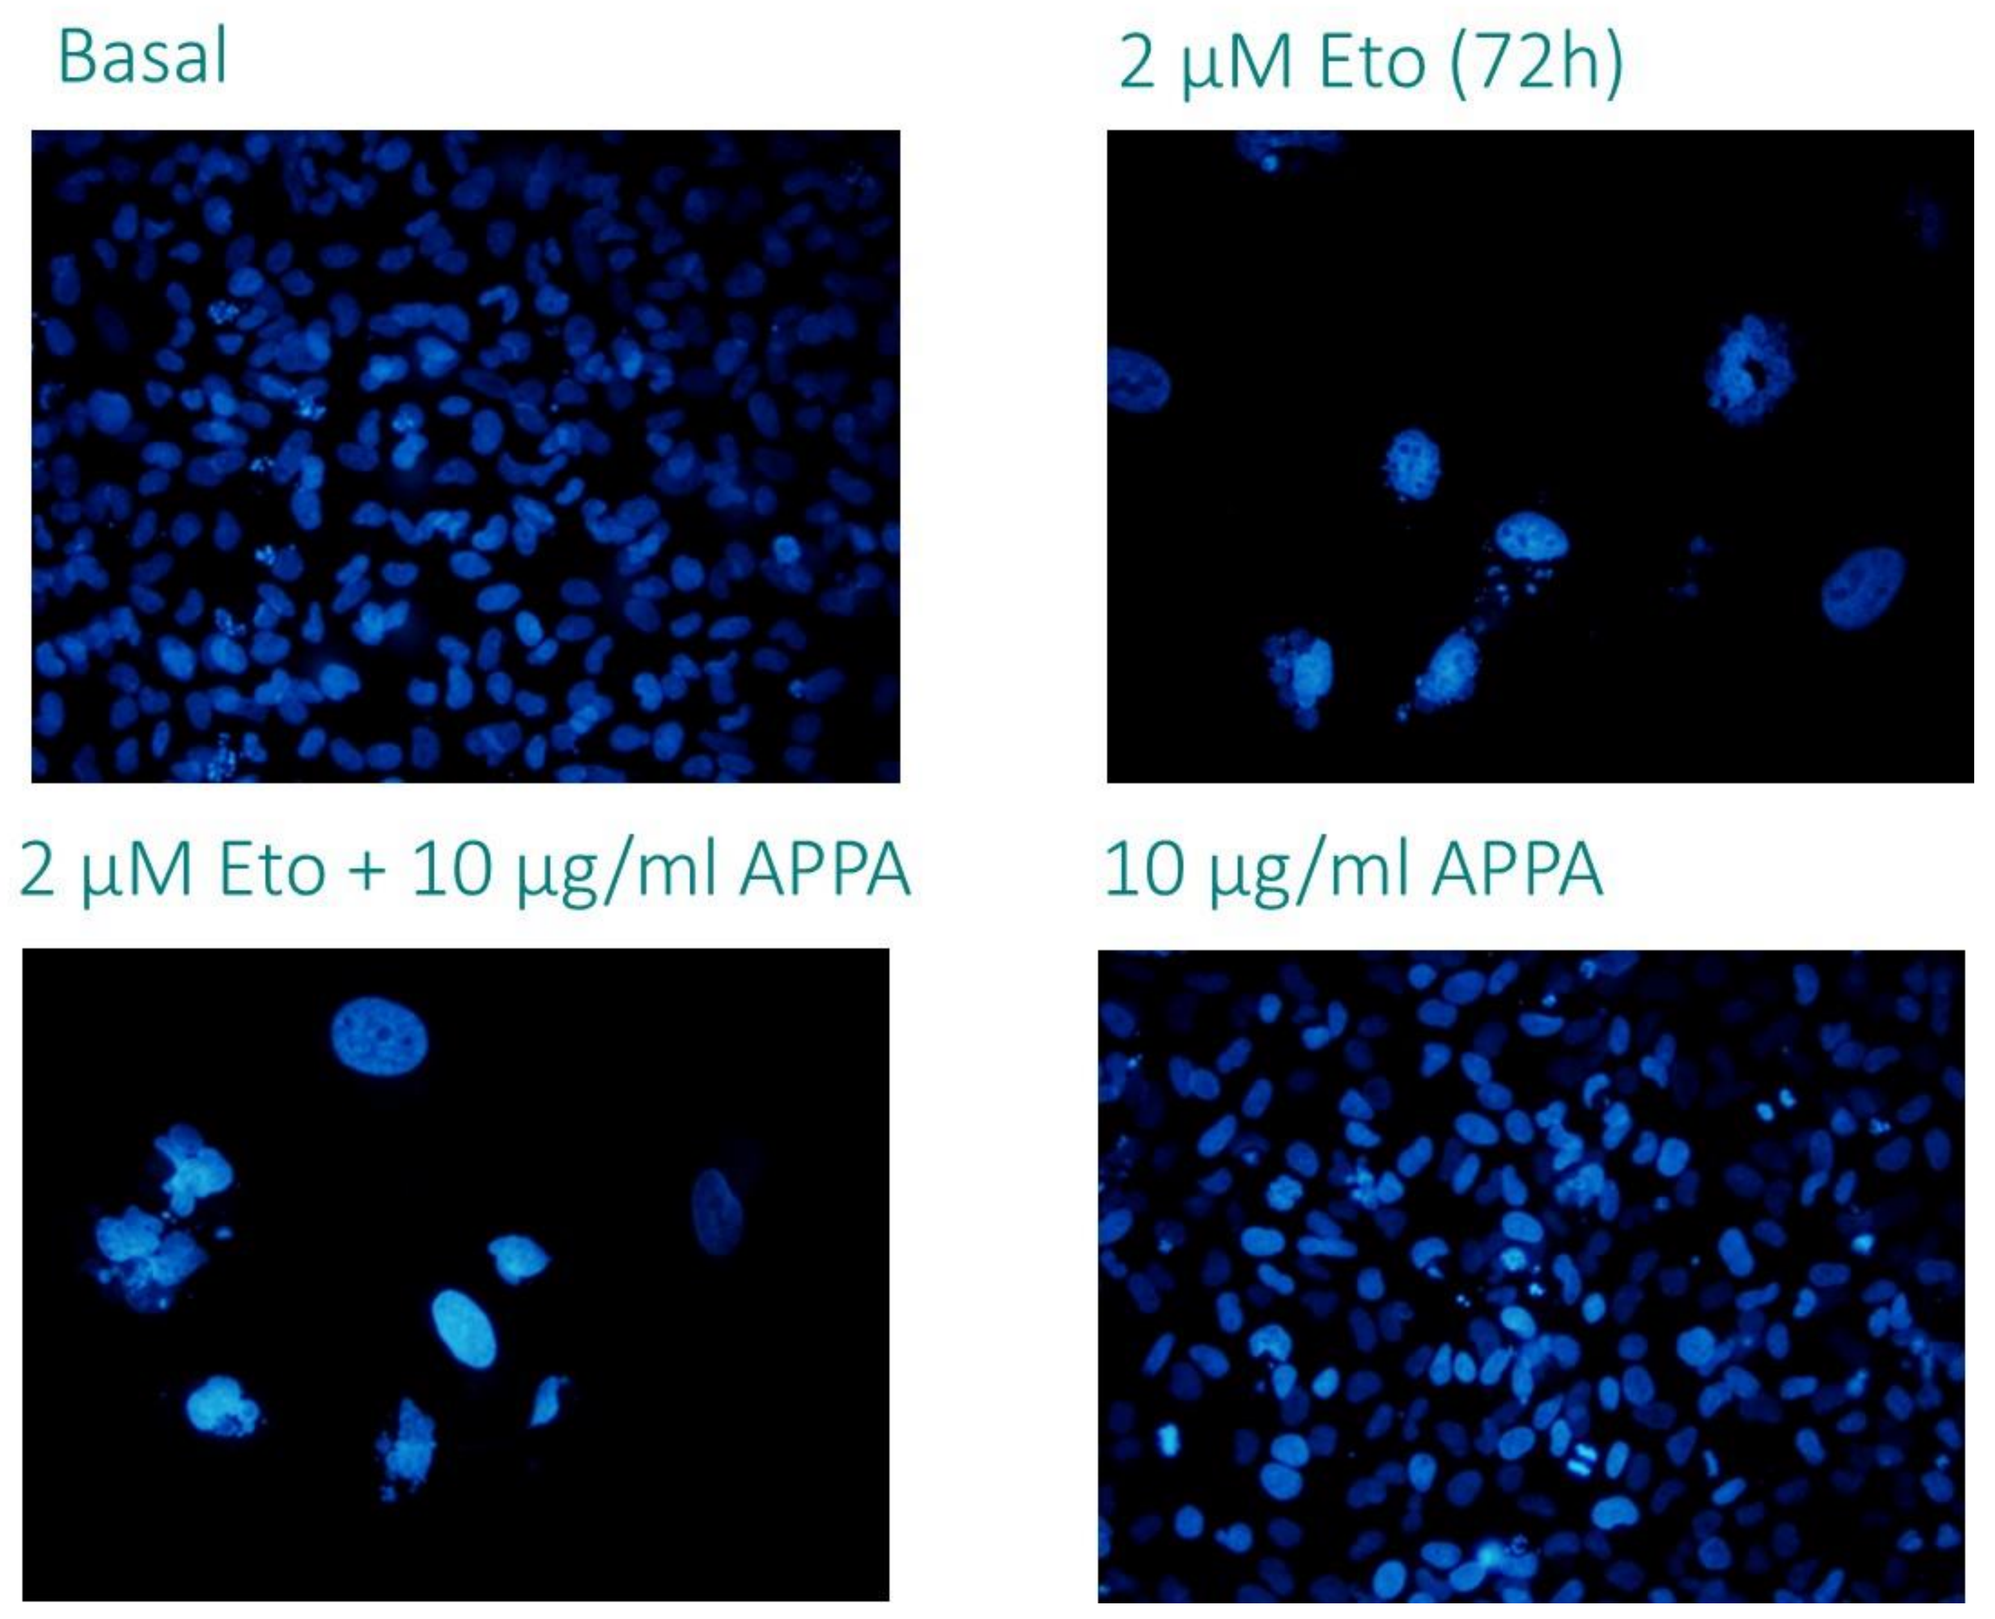
Basal
2 µM Eto (72h)
2 µM Eto + 10 µg/ml APPA
10 µg/ml APPA

B

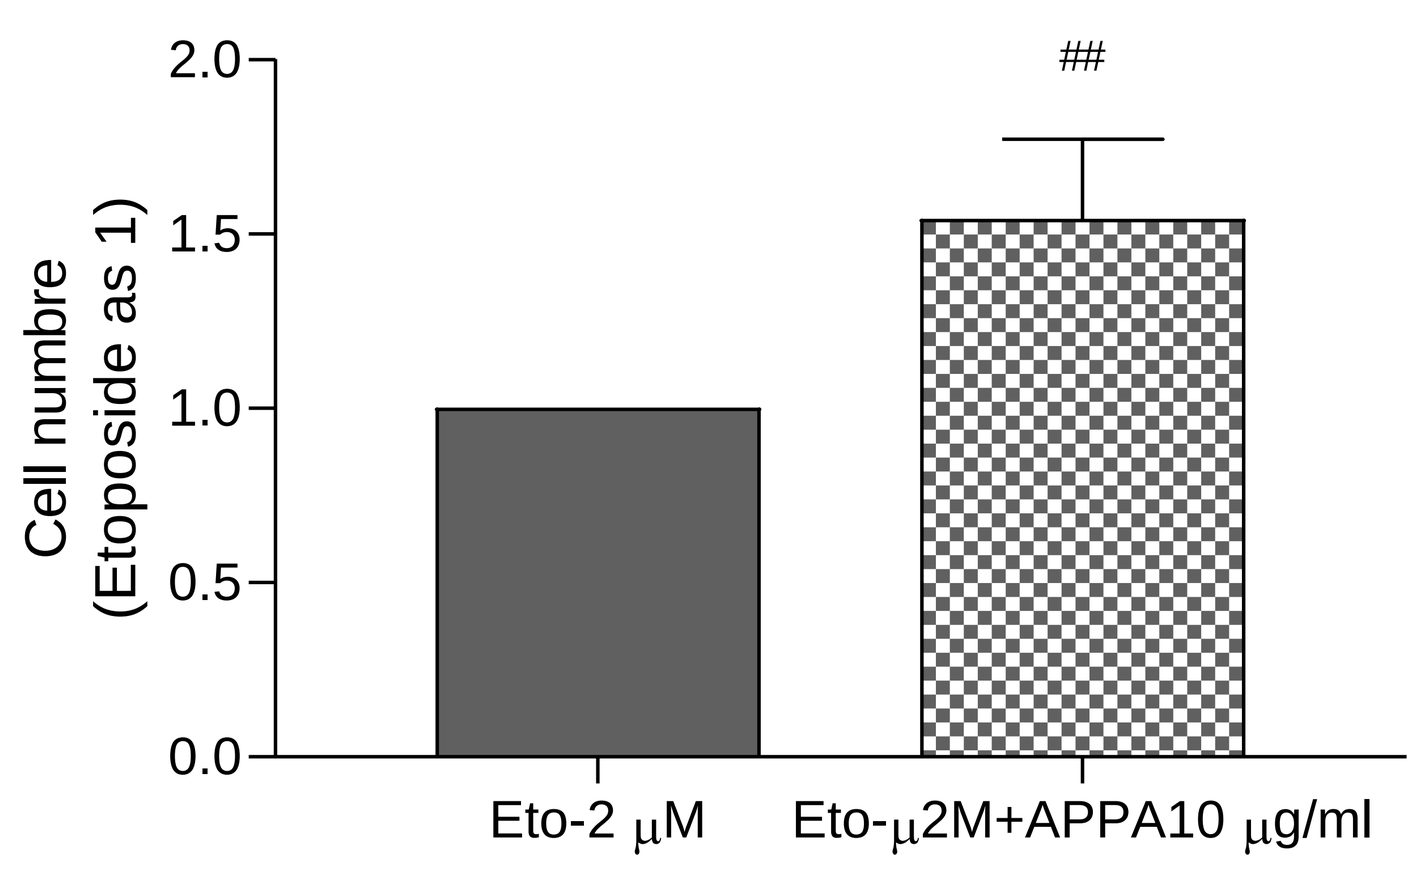
Cell numbre
(Etoposide as 1)
2.0
1.5
1.0
0.5
0.0
##
Eto-2 µM
Eto-µ2M+APPA10 µg/ml

Figure 2. Senolitic index. A) Representative image corresponding to nucleic from cells in basal condition, incubated in presence of 2 µM Etoposide, in combination with 2 µM Etoposide + 10 µg/ml APPA, and with only 10 µg/ml APPA. **B)** Quantification of nucleic presents in each condition relative to Etoposide. ## p<0.01.

The analysis of apoptotic cells after stimulation with Etoposide 2 µM or **APPA 10 µg/ml did not show any modulation of apoptosis** in any of the conditions evaluated (**Figure 3**). These data suggests that APPA, at least in this chondrocyte cell line (TC/28a2) could have senomorphic effects.

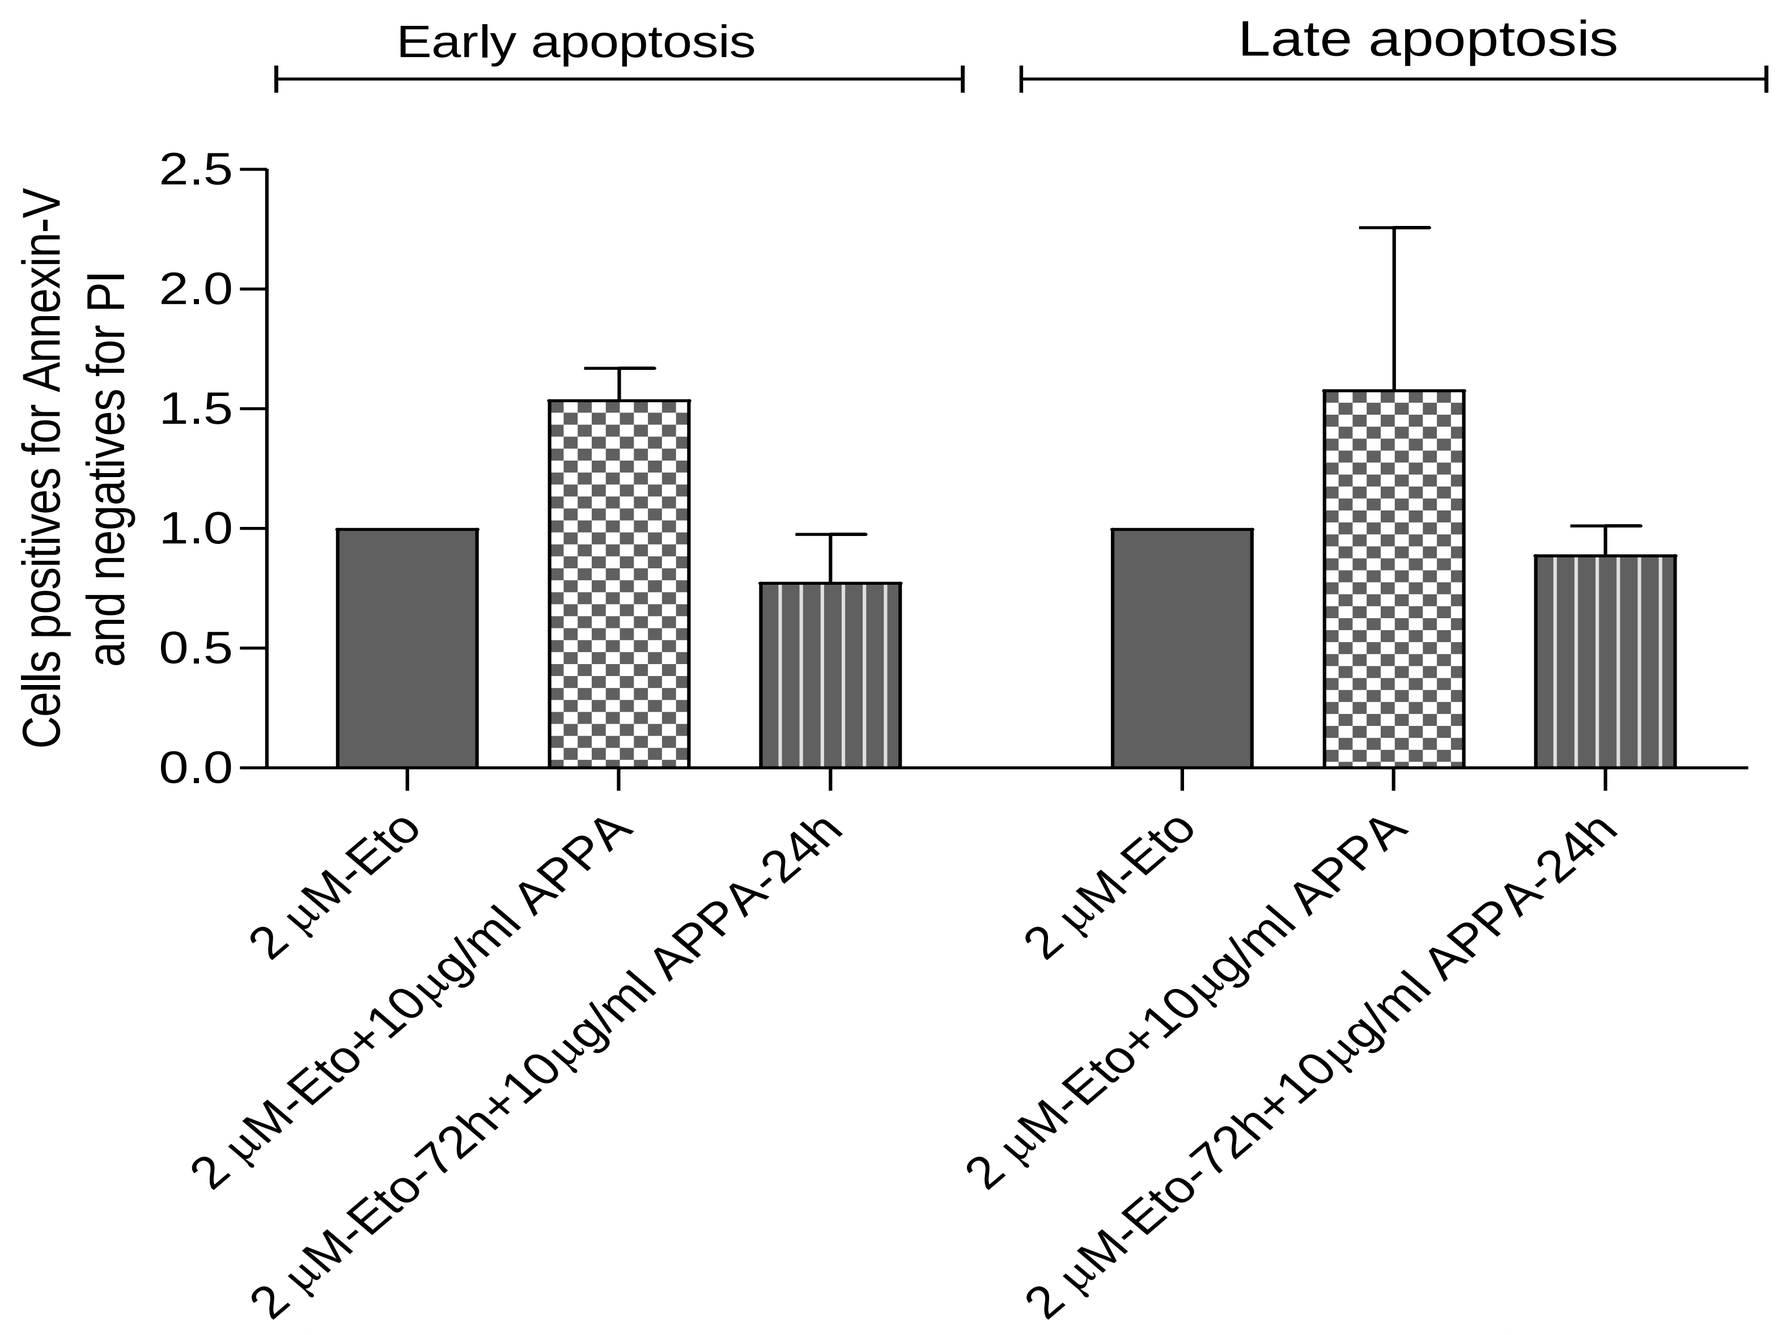
Early apoptosis
Late apoptosis
Cells positives for Annexin-V
and negatives for PI
2.5
2.0
1.5
1.0
0.5
0.0
2 µM-Eto
2 µM-Eto+10µg/ml APPA
2 µM-Eto-72h+10µg/ml APPA-24h
2 µM-Eto
2 µM-Eto+10µg/ml APPA
2 µM-Eto-72h+10µg/ml APPA-24h

Figure 3. Analysis of Apoptotic cells expressed relative to positive stimuli (2µM Eto during 72h).

These results were partially replicated in **human articular OA chondrocytes**, where APPA decreased the ß-galactosidase activity (**Figure 4-A, Table-1**) as well as *CDKN1A* gene expression levels induced by Etoposide (**Figure 4-B, Table 1**). However, APPA did not increase the total number of cells. The low proliferation capacity of OA chondrocytes from elderly patients may be an explanation for this result.

A

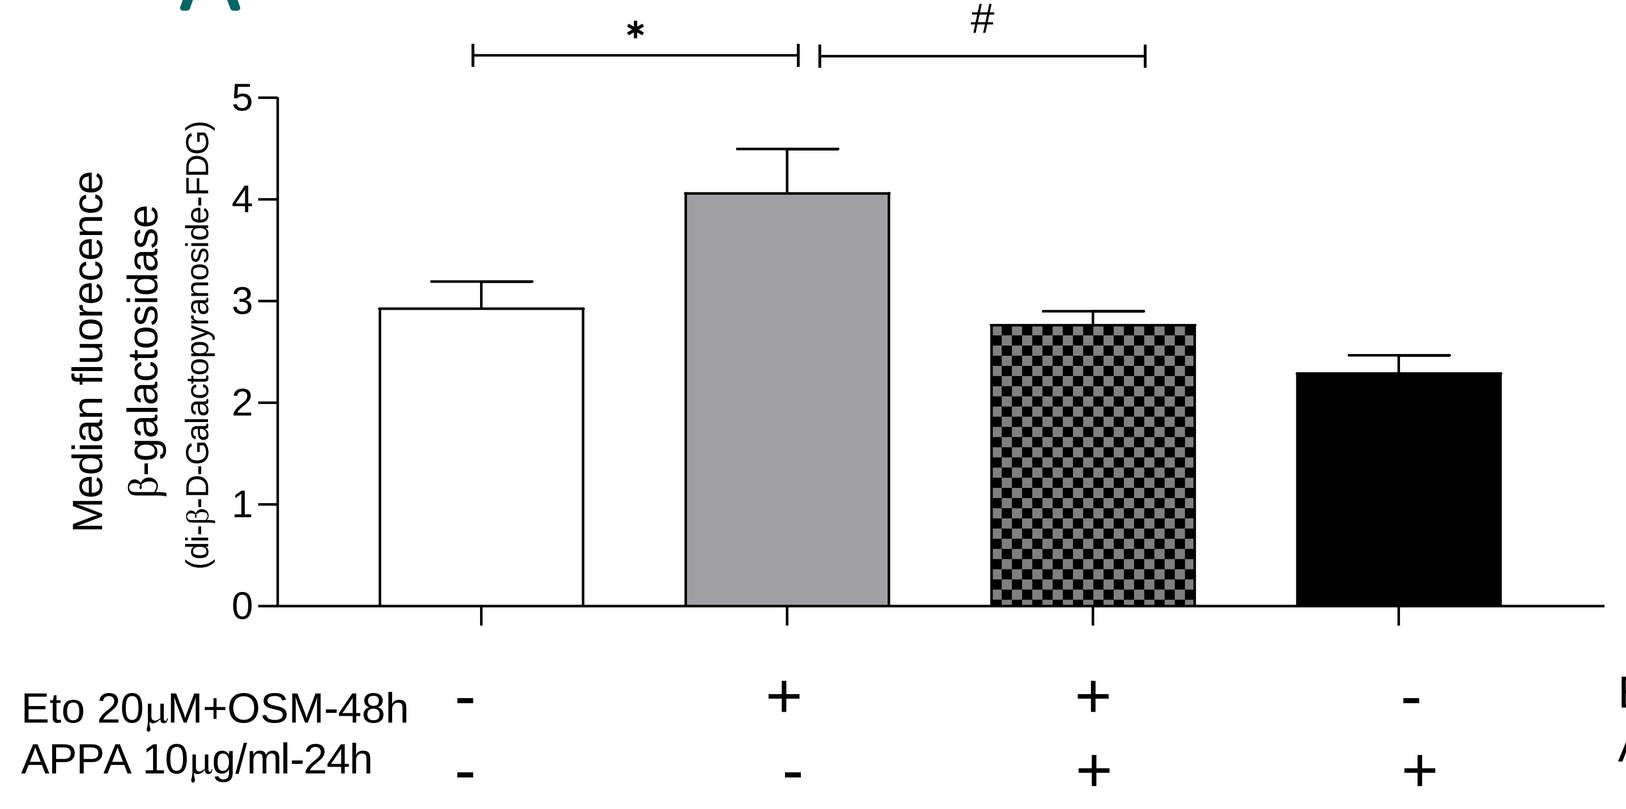
Median fluorecence
β-galactosidase
(di-β-D-Galactopyranoside-FDG)
5
4
3
2
1
0
*
#
Eto 20µM+OSM-48h - + + -
APPA 10µg/ml-24h - - + +

B

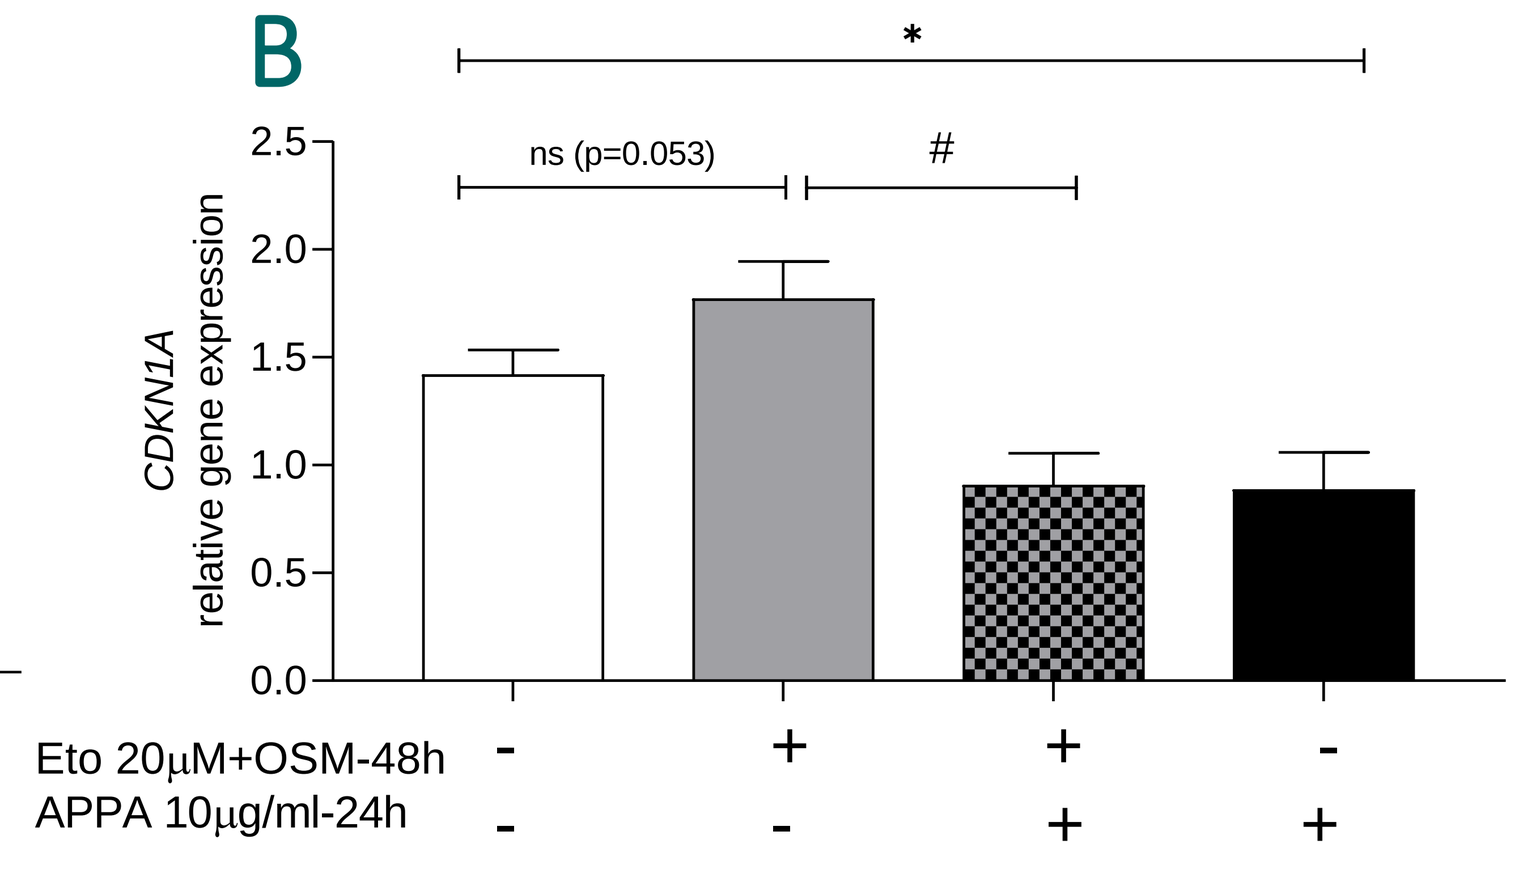
CDKN1A
relative gene expression
2.5
2.0
1.5
1.0
0.5
0.0
*
ns (p=0.053)
#
Eto 20µM+OSM-48h - + + -
APPA 10µg/ml-24h - - + +

Figure 4. APPA effect on human articular chondrocytes A) ß-Galactosidase activity, **B)** *CDKN1A* gene expression. *p<0.05, #p<0.05.

Table 1. APPA effect on human articular chondrocytes. APPA decreases the ß-galactosidase activity as well as CDKN1A gene expression levels induced by 20 µM Eto.

	Basal	Eto (20 µM + OSM 10 ng/ml-48h)	p value (Mann Whitney)
ß-Galactosidase activity (FDG)	2.931 ± 0.263	4.064 ± 0.435	* (p=0.037)
CDKN1A gene expression	1.418 ± 0.11	1.77 ± 0.17	0.053

	Eto (20 µM + OSM 10 ng/ml-48h)	Eto (20 µM-48h)+APPA (10 µg/ml)-24h	p value (Mann Whitney)
ß-Galactosidase activity (FDG)	4.064 ± 0.435	2.77 ± 0.132	# (p=0.018)
CDKN1A gene expression	1.77 ± 0.17	0.9058 ± 0.14	# (p=0.042)

Data represented in the table corresponding to mean±SEM, N=8

Conclusion APPA reduced the number of senescent cells and increased the number of cells without causing apoptosis, which indicates that APPA may be senomorphic.